Comparison of cfDNA Reference Material Prepared using Enzymatic Fragmentation or Sonication for the Validation of Liquid Biopsy Assays

Aldo Mele[1], Hannah Child[1], Katarzyna Wilczynska[1], Christian Paar[2] Simi Chacko[3] and Julie Wickenden[1]

1 Horizon Discovery, United Kingdom 2 Kepler University Hospital, Austria 3 Atlantic Cancer Research Institute, Canada

Introduction

Liquid biopsies hold great promise to revolutionize the field of clinical oncology testing. Cell-free DNA (cfDNA) can be extracted from a routine patient blood sample and used to determine the genetic profile of a solid tumor located elsewhere within the body. This facilitates more informative disease management for the clinician, without the need for invasive surgery for the patient. With new cfDNA NGS assays being able to detect variants from as little as 2-10ng DNA, assay validation to ensure sufficient accuracy has never been so critical. Reference materials that closely mimic real cfDNA samples are essential to support this effort. Here we present results from a comparative study of DNA fragmentation methods applied during the production of cfDNA reference standards. We show a comparison between enzymatic fragmentation and mechanical shearing (sonication), and the benefit of including a size selection step for data accuracy and performance of NGS gene panel workflow.

Methods

DNA extracted from engineered cancer cell lines, representing the Multiplex 1 blend at 5% or 0.1%, was fragmented by mechanical or enzymatic shearing. In addition, a size selection step was included to obtain a fragment size distribution profile that closely mimics real cfDNA samples. The allele frequency of specific variants was confirmed by ddPCR. The eight-sample cfDNA material experimental set was externally tested on the Illumina TruSight Tumor15 (TST-15) panel and the Oncomine Breast cfDNA Assay v2 (OBA v2) to assess library preparation and variant calling performance.

NGS: Sequencing was performed on the MiSeqDx system in RUO mode and the Ion S5 for the TST-15 and OBA v2 assays respectively. MiSeqDx system filter settings for analysis with Variant Studio (and automatic analysis) were: Read depth >500 and MAF >2%.

Results

Tapestation analysis confirmed that both sonication and enzymatic fragmentation produced cfDNA with a peak size in line with real clinical samples (Fig 1). Proof of principle variant detection by ddPCR confirmed the presence of three of the eight mutations across 2 of four genes (EGFR, KRAS, NRAS and PIK3CA) at either 0.1% or 5% variant allele frequency (Fig 2). Library preparation using both Illumina TST-15 gene panel and OBA v2 showed good library yield across all eight samples (Fig 3). NGS sequencing results showed good and comparable variant calling ability between both sonicated and enzymatically sheared samples (Fig 4). Whilst enzymatically sheared cfDNA did not show a significant increase in NGS library yield, it did display slightly more accurate variant calling on the TST-15 assay, in addition to a more defined tapestation profile – centred around 168bp, which was further enhanced when combined with a size selection step (Fig 1A yellow trace, Fig 3 and Fig 4). In addition, due to a lower limit of detection, the OBA v2 was able to detect many of the variants at 0.1% allele frequency (Fig 4).

A)

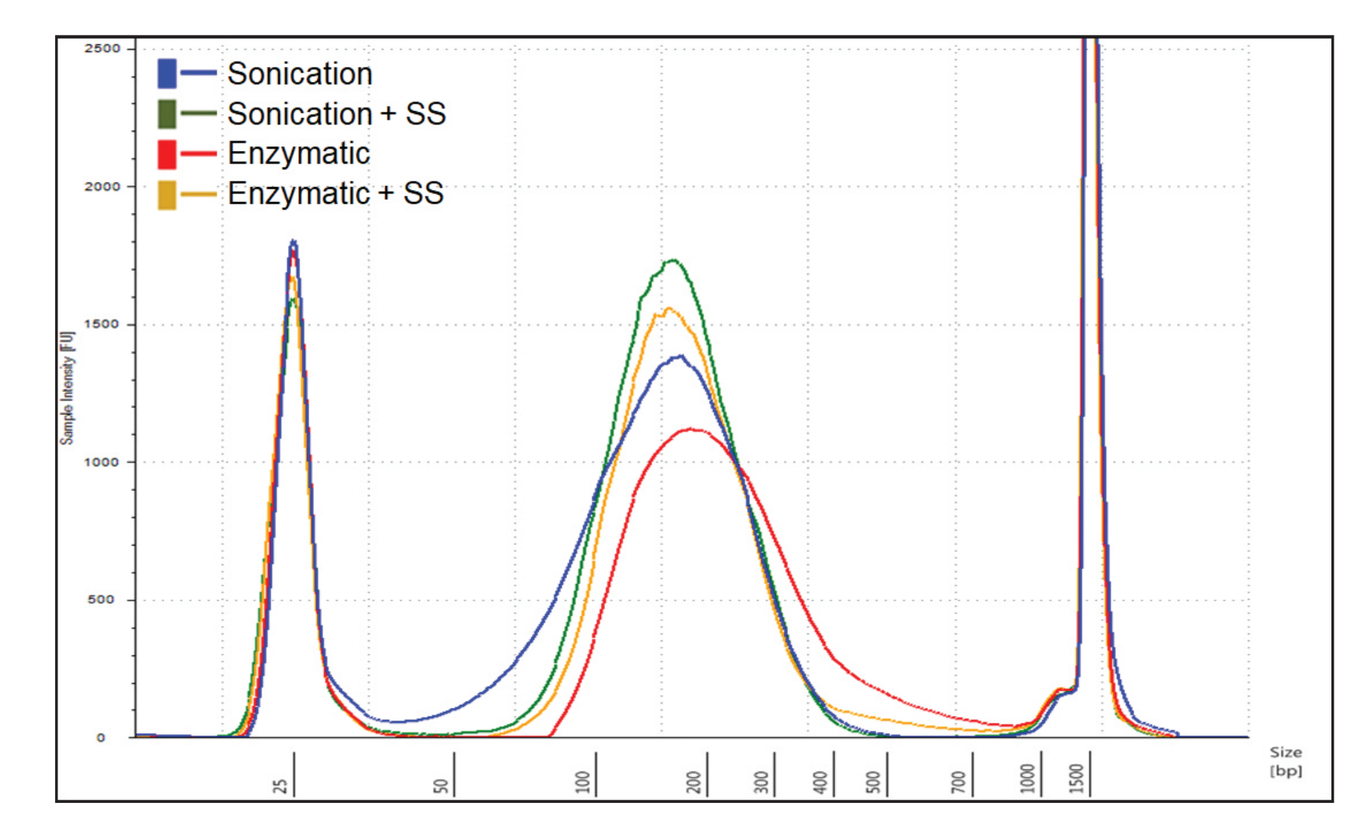

Sonication
Sonication + SS
Enzymatic
Enzymatic + SS

B)

Sample	Peak (bp)	Region [35-950bp] Average Fragment Length (bp)	Region [35-950bp] % of Total
Sonication	178	170	95.08
Sonication + SS	171	180	96.54
Enzymatic	187	243	94.95
Enzymatic + SS	168	196	95.58

Figure 1: Size Distribution
(A) TapeStation analysis of size distribution of all sample conditions (B) Table listing peak size and average fragment length

	Mutation	5% AF samples Expected AF (%)	5% AF samples Acceptance Criteria (%)	5% AF samples Measured AF (%)	0.1% AF samples Expected AF (%)	0.1% AF samples Acceptance Criteria (%)	0.1% AF samples Measured AF (%)
Sonication	EGFR L858R	5	3.5 - 6.5	4.5	0.10	0.05-0.15	0.13
	EGFR V769-D770insASV	5	3.5 - 6.5	4.4	0.10	0.07 - 0.20	0.11
	NRAS Q61K	6.3	4.4 - 8.2	6.3	0.13	0.07-0.20	0.14
Sonication + SS	EGFR L858R	5	3.5 - 6.5	4.6	0.10	0.05-0.15	0.13
	EGFR V769-D770insASV	5	3.5 - 6.5	4.2	0.10	0.07 - 0.20	0.10
	NRAS Q61K	6.3	4.4 - 8.2	6.4	0.13	0.07-0.20	0.12
Enzymatic	EGFR L858R	5	3.5 - 6.5	4.7	0.10	0.05-0.15	0.12
	EGFR V769-D770insASV	5	3.5 - 6.5	4.5	0.10	0.07 - 0.20	0.10
	NRAS Q61K	6.3	4.4 - 8.2	6.6	0.13	0.07-0.20	0.13
Enzymatic + SS	EGFR L858R	5	3.5 - 6.5	4.7	0.10	0.05-0.15	0.13
	EGFR V769-D770insASV	5	3.5 - 6.5	4.3	0.10	0.07 - 0.20	0.10
	NRAS Q61K	6.3	4.4 - 8.2	6.5	0.13	0.07-0.20	0.14

Figure 2: Representative ddPCR QC analysis on three of the eight mutations at either 0.1% or 5% variant AF

	TruSight Tumor 15	ng/µL
1	5% sonic. _ 31097	27.6
2	5% sonic. + SS _ 31168	49.3
3	5% enzymatic _ 31116	44.7
4	5% enzymatic + SS _ 31119	38.5
5	0,1% sonic. _ 29637	50.5
6	0,1% sonic. + SS _ 31169	49.7
7	0,1% enzymatic _ 31117	42.7
8	0,1% enzymatic + SS _31120	57.2
	Oncomine Breast cfDNA Assay v2	**nM**
1	5% sonic. _ 31097	11.3
2	5% sonic. + SS _ 31168	6.8
3	5% enzymatic _ 31116	11.5
4	5% enzymatic + SS _ 31119	10.0
5	0,1% sonic. _ 29637	7.7
6	0,1% sonic. + SS _ 31169	7.9
7	0,1% enzymatic _ 31117	10.4
8	0,1% enzymatic + SS _31120	12.2

Figure 3: NGS Library Yield

Expected Variant AF (%) (ddPCR)	Mutations Gene	Variant	Type	Protein Position	AA Change	Exon	Consequence	Sonication (31097) TST15	Sonication (31097) Oncomine	Sonication + Size Selection (31168) TST15	Sonication + Size Selection (31168) Oncomine	Enzymatic (31116) TST15	Enzymatic (31116) Oncomine	Enzymatic + Size Selection (31119) TST15	Enzymatic + Size Selection (31119) Oncomine
5.0	EGFR	ΔE746 - A750*	deletion	0	KELREA/K	19/28	inframe-deletion	7.1	-	8.2	-	4.7	-	5.5	-
5.0	EGFR	L858R	snv	858	L/R	21/28	missense	4.4	3.9	2.7	4.7	5.9	4.4	6.1	5.2
5.0	EGFR	T790M*	snv	790	T/M	20/28	missense	5.6	-	3.6	-	5.9	-	4.2	-
5.0	EGFR	V769 - D770insASV*	insertion	766	M/MASV	20/28	inframe-insertion	5.9	-	4.0	-	4.3	-	3.8	-
6.3	KRAS	G12D	snv	12	G/D	2/6	missense	7.7	6.5	7.9	6.6	6.5	7.2	5.8	7.0
6.3	NRAS	Q61K*	snv	61	Q/K	3/7	missense	9.2	-	5.2	-	6.5	-	6.8	-
6.3	NRAS	A59T*	snv	59	A/T	3/7	missense	7.0	-	8.3	-	6.2	-	6.9	-
6.3	PIK3CA	E545K	snv	545	E/K	10/21	missense	5.3	6.7	6.3	5.9	6.5	5.6	6.7	7.0

Expected Variant AF (ddPCR)	Mutations Gene	Variant	Type	Protein Position	AA Change	Exon	Consequence	Sonication (31097) TST15	Sonication (31097) Oncomine	Sonication + Size Selection (31168) TST15	Sonication + Size Selection (31168) Oncomine	Enzymatic (31116) TST15	Enzymatic (31116) Oncomine	Enzymatic + Size Selection (31119) TST15	Enzymatic + Size Selection (31119) Oncomine
0.1%	EGFR	ΔE746 - A750*	deletion	0	KELREA/K	19/28	inframe-deletion	Not detected *(below LOD)*	-	Not detected *(below LOD)*	-	Not detected *(below LOD)*	-	Not detected *(below LOD)*	-
	EGFR	L858R	snv	858	L/R	21/28	missense		0.11		-		-		-
	EGFR	T790M*	snv	790	T/M	20/28	missense		-		-		-		-
	EGFR	V769 - D770insASV*	insertion	766	M/MASV	20/28	inframe-insertion		-		-		-		-
	KRAS	G12D	snv	12	G/D	2/6	missense		0.31		0.12		0.12		0.13
	NRAS	Q61K*	snv	61	Q/K	3/7	missense		-		-		-		-
	NRAS	A59T*	snv	59	A/T	3/7	missense		-		-		-		-
	PIK3CA	E545K	snv	545	E/K	10/21	missense		0.19		0.23		0.13		0.17

** Variants not present on Oncomine Breast cfDNA Assay v2*

Figure 4: NGS results from 5% and 0.1% AF cfDNA test samples run on both the Illumina TST-15 assay and Oncomine Breast cfDNA Assay v2

Conclusion

Results show good performance of both sonicated and enzymatically sheared cfDNA material, where all variants present above the LOD could be detected on the Illumina TST-15 assay.

Whilst enzymatically sheared cfDNA did not show any increase in NGS library yield, it did display slightly more accurate variant calling, in addition to a more defined tapestation profile – centred around 168bp, which was further enhanced when combined with a size selection step (Fig 1A yellow trace).

This highlights the potential of these alternative techniques to produce cfDNA that is highly commutable to patient samples and suitable for the validation of ddPCR and NGS liquid biopsy assays.

In partnership with:

INSTITUT ATLANTIQUE DE RECHERCHE SUR LE CANCER ATLANTIC CANCER RESEARCH INSTITUTE

Kepler Universitäts Klinikum

www.horizondiscovery.com

©2018 Horizon Discovery Group Company—All rights reserved. UK Registered Head Office: Building 8100, Cambridge Research Park, Cambridge, CB25 9TL, United Kingdom. UK Registration No. 05363294 | UK VAT No. GB 189 1993 44